RAP-219: A Differentiated ASM Targeting Restricted Brain Expression of γ8 Transmembrane AMPA Receptor Regulatory Protein (TARPγ8)

Poster 3.355

Stephen Greene, PharmD[1]; Marie Cohilis, PhD[2]; Michel Koole, PhD[2]; Kim Serdons, PhD[2]; Marissa Herbots, MSN[3]; Kathleen Sierens, MD[3]; Jan de Hoon, PhD, MD[3]; Koen Van Laere, MD, PhD[2]; Swamy Yeleswaram, PhD[1]

[1]Rapport Therapeutics, Inc., Boston, MA, USA; [2]Nuclear Medicine and Molecular Imaging, Department of Imaging and Pathology, Katholiek Universiteit Leuven, Leuven, Belgium; [3]Center for Clinical Pharmacology, Department of Pharmaceutical and Pharmacological Sciences, University of Leuven, Leuven, Belgium.

Background

- Transmembrane AMPA receptor regulatory proteins (TARPs) are anatomically restricted AMPA receptor accessory proteins that control gating and function[1]
- In preclinical studies, TARPγ8 was highly expressed in the cortex, hippocampus, and amygdala,[2] key regions of the brain involved in the initiation and propagation of seizures
- RAP-219, a potent and selective TARPγ8 negative allosteric modulator, offers a precision approach for the treatment of focal epilepsy via targeted AMPA receptor inhibition
- In this study, we quantified the neuroanatomic expression of TARPγ8 in humans and evaluate the relationship between receptor occupancy (RO) and pharmacokinetics (PK) of RAP-219

Methods

- Healthy adults (18-55 y) were enrolled in 3 cohorts of an open-label study and received an oral dose of RAP-219 once daily for 14 days following ≥10 h of fasting

Figure 1. PET Study RAP-219 Dose Cohorts

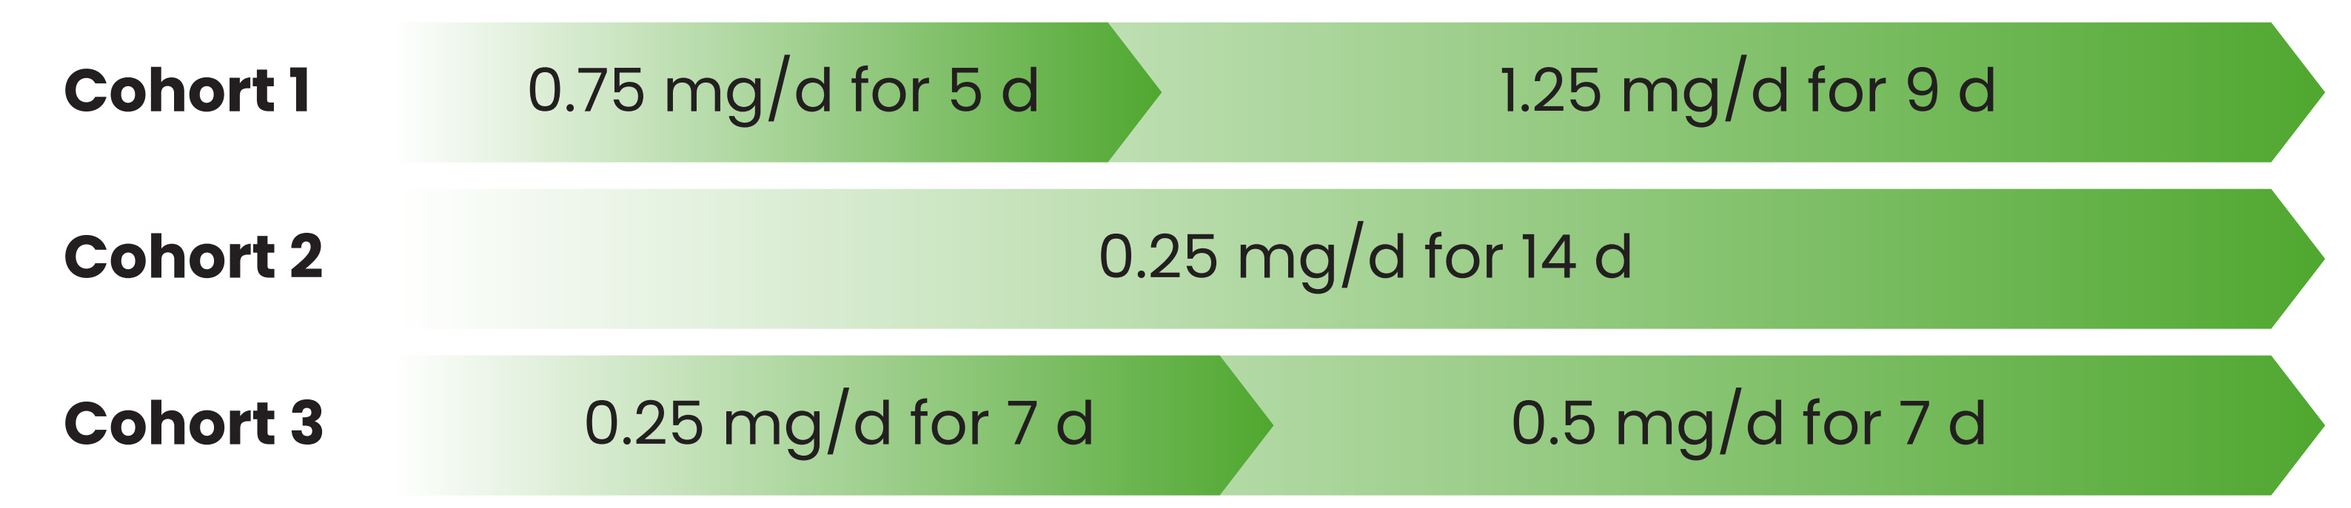
Cohort 1
0.75 mg/d for 5 d
1.25 mg/d for 9 d
Cohort 2
0.25 mg/d for 14 d
Cohort 3
0.25 mg/d for 7 d
0.5 mg/d for 7 d

- Pre-dose baseline (D1) and post-dose (D15; 3-4 h after RAP-219 administration) PET scans were obtained using an intravenous injection of a TARPγ8-specific tracer, [18F]JNJ-64511070 (185 MBq [5 mCi] ±10%), and arterial blood sampling
 - Baseline PET scans were used to evaluate TARPγ8 expression; post-dose PET scans were compared to baseline to assess RAP-219 on TARPγ8 RO
 - Serial blood samples for PK were collected on D2 and D15
- PK and adverse events (AEs; NCI-CTCAE v5.0 grading: Grades 1 [mild] to 5 [AE-related death]) were summarized by cohort overall
- TARPγ8 RO was estimated using baseline and post-dose distribution volume and Lassen plots, assuming constant non-displaceable distribution volume between baseline and post-dose periods
 - Distribution volumes were determined by a two-tissue compartment model
 - Correlation between RAP-219 plasma concentrations (Cp; D15, 4 h post-dose) and estimated RO was determined using a sigmoid E_{max} model
 - $E(\text{Occupancy} \mid \text{PK plasma}) = E_{max} \times (\text{PK plasma}^h/[\text{PK plasma}^h + EC_{50}{}^h])$

Results

- Sixteen subjects (mean age, 39 ± 11.1 y; 93.8% male) were sequentially enrolled into Cohort 1 (n=6), Cohort 2 (n=6), and Cohort 3 (n=4)

Table 1. Subject Demographics

	Cohort 1 n=6	Cohort 2 n=6	Cohort 3 n=4
Male, n (%)	6 (100)	5 (83.3)	4 (100)
Age, years, mean ± SD	29.8 ± 8.4	46.2 ± 5.3	42.0 ± 12.8
Race, n (%)			
White	6 (100)	6 (100)	4 (100)

TARPγ8 Expression

- The greatest mean distribution volume of TARPγ8 at baseline was in the neocortex and mesial temporal lobe, including the hippocampus and amygdala (**Figures 2A** and **5A**)
- RAP-219 (14-day treatment) blocked tracer binding at Day 15 in post-dose scans (**Figures 2B** and **5B**)

Figure 2. Average TARPγ8 Volume of Distribution Across Brain Regions at A. Baseline (D1) and B. After RAP-219 Dose (D15)

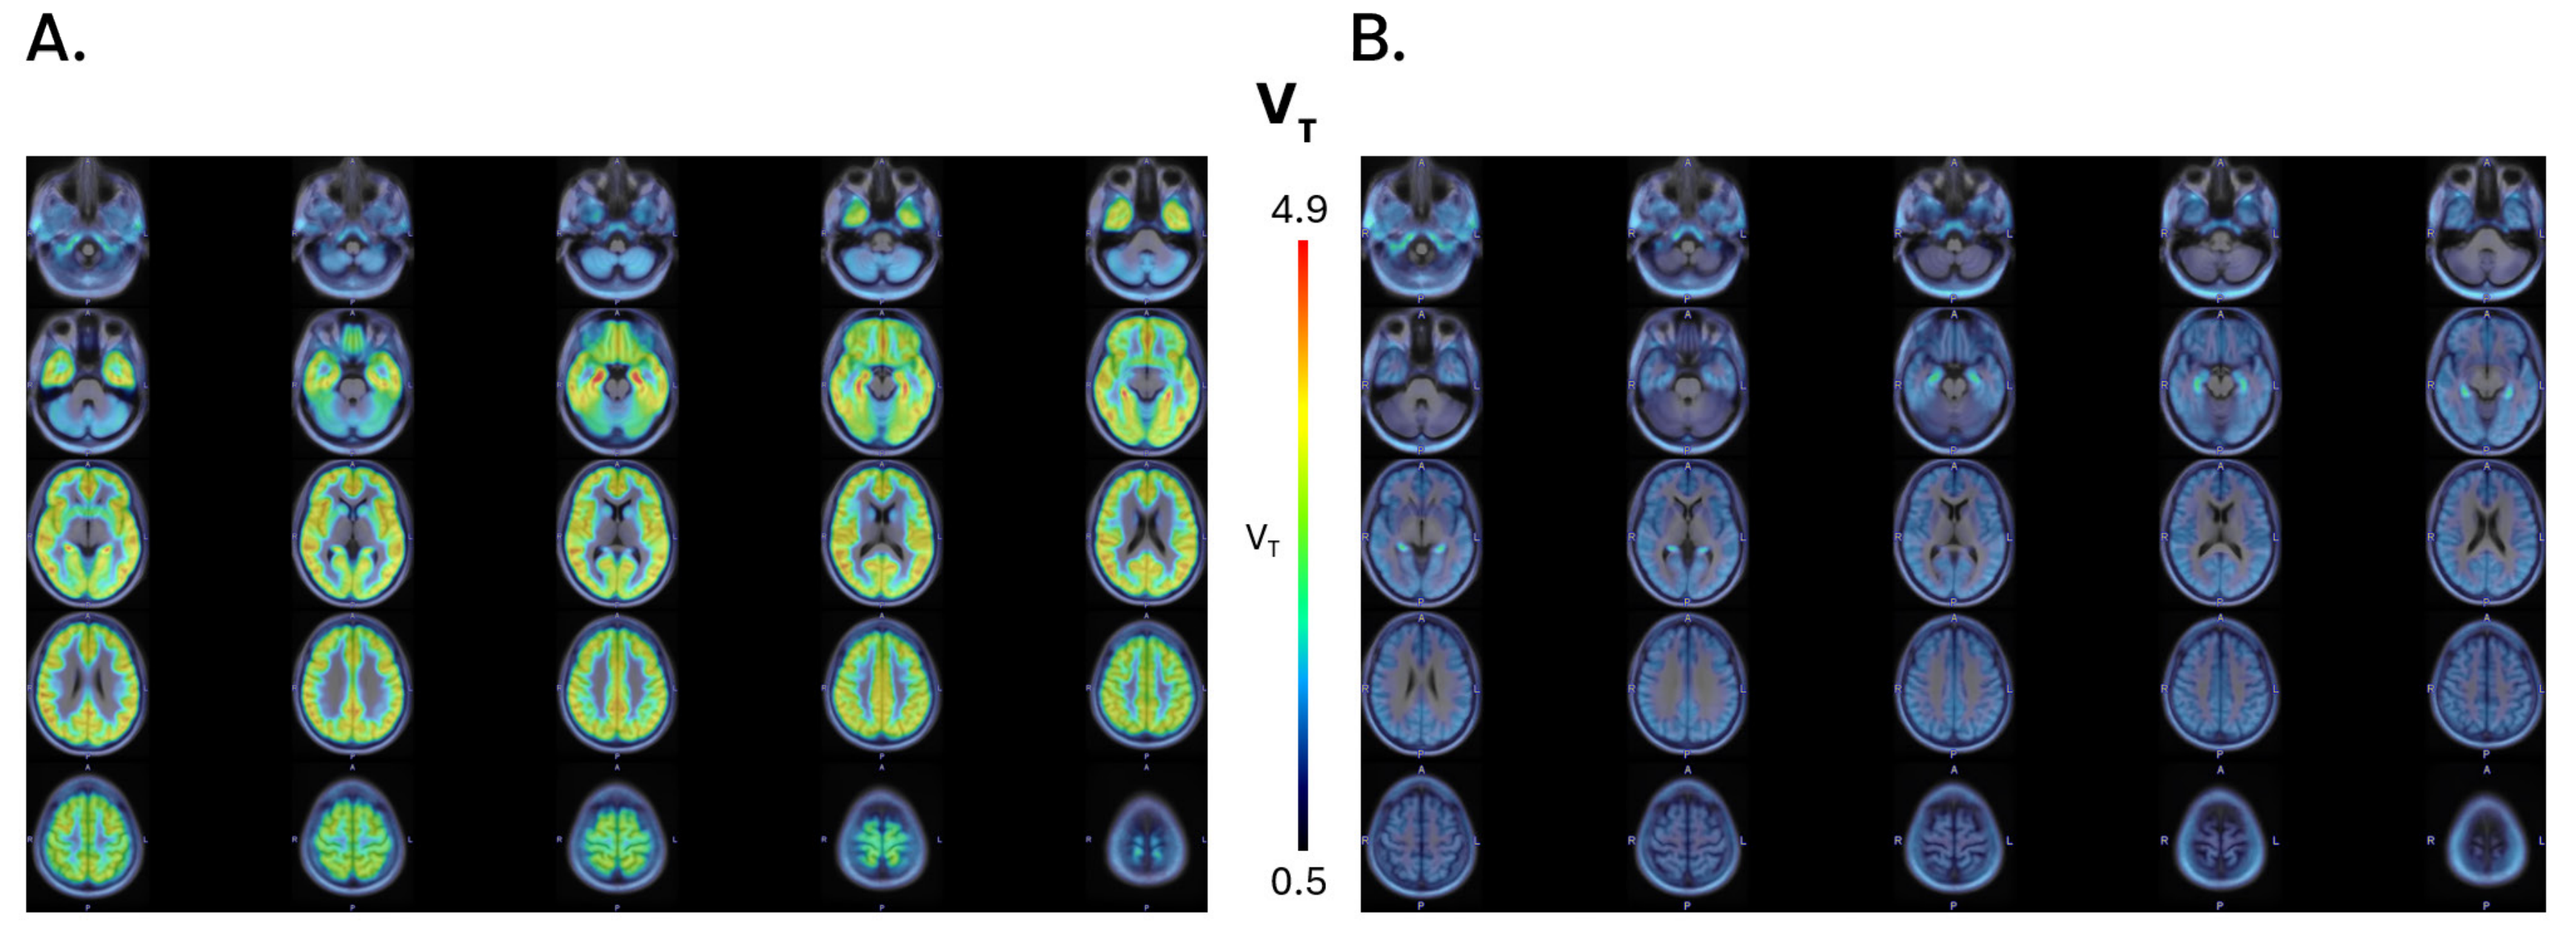
A.
B.
V_T
4.9
0.5

V_T is superposed to a T1-weighted MRI.
MRI - magnetic resonance imaging; V_T – volume of distribution.

Pharmacokinetics

- The PK profile observed was mainly consistent with previous multiple-dose studies
 - $AUC_{0\text{-}t}$ on D2 was 22.9 h*ng/mL in Cohort 1; in Cohorts 2 and 3, $AUC_{0\text{-}t}$ was approximately 7 h*ng/mL
 - RAP-219 exhibited an increase in C_{max} from D2 (median C_{max} [ng/mL]: Cohort 1, 2.03; Cohort 2, 0.816; Cohort 3, 0.705) to D15 (median C_{max} [ng/mL]: Cohort 1, 9.18; Cohort 2, 2.33; Cohort 3: 4.18) across all cohorts
 - A median T_{max} of 2 h was observed in all cohorts on D2 , which increased to 2 h, 1.99 h, and 5.95 h on D15 in Cohorts 1, 2, and 3, respectively
 - On D15, the mean elimination half-life of RAP-219 was 525 h in Cohort 1, 670 h in Cohort 2, and 706 h in Cohort 3

Figure 3. Mean RAP-219 Plasma Concentration Following RAP-219 Adminstration (D15)

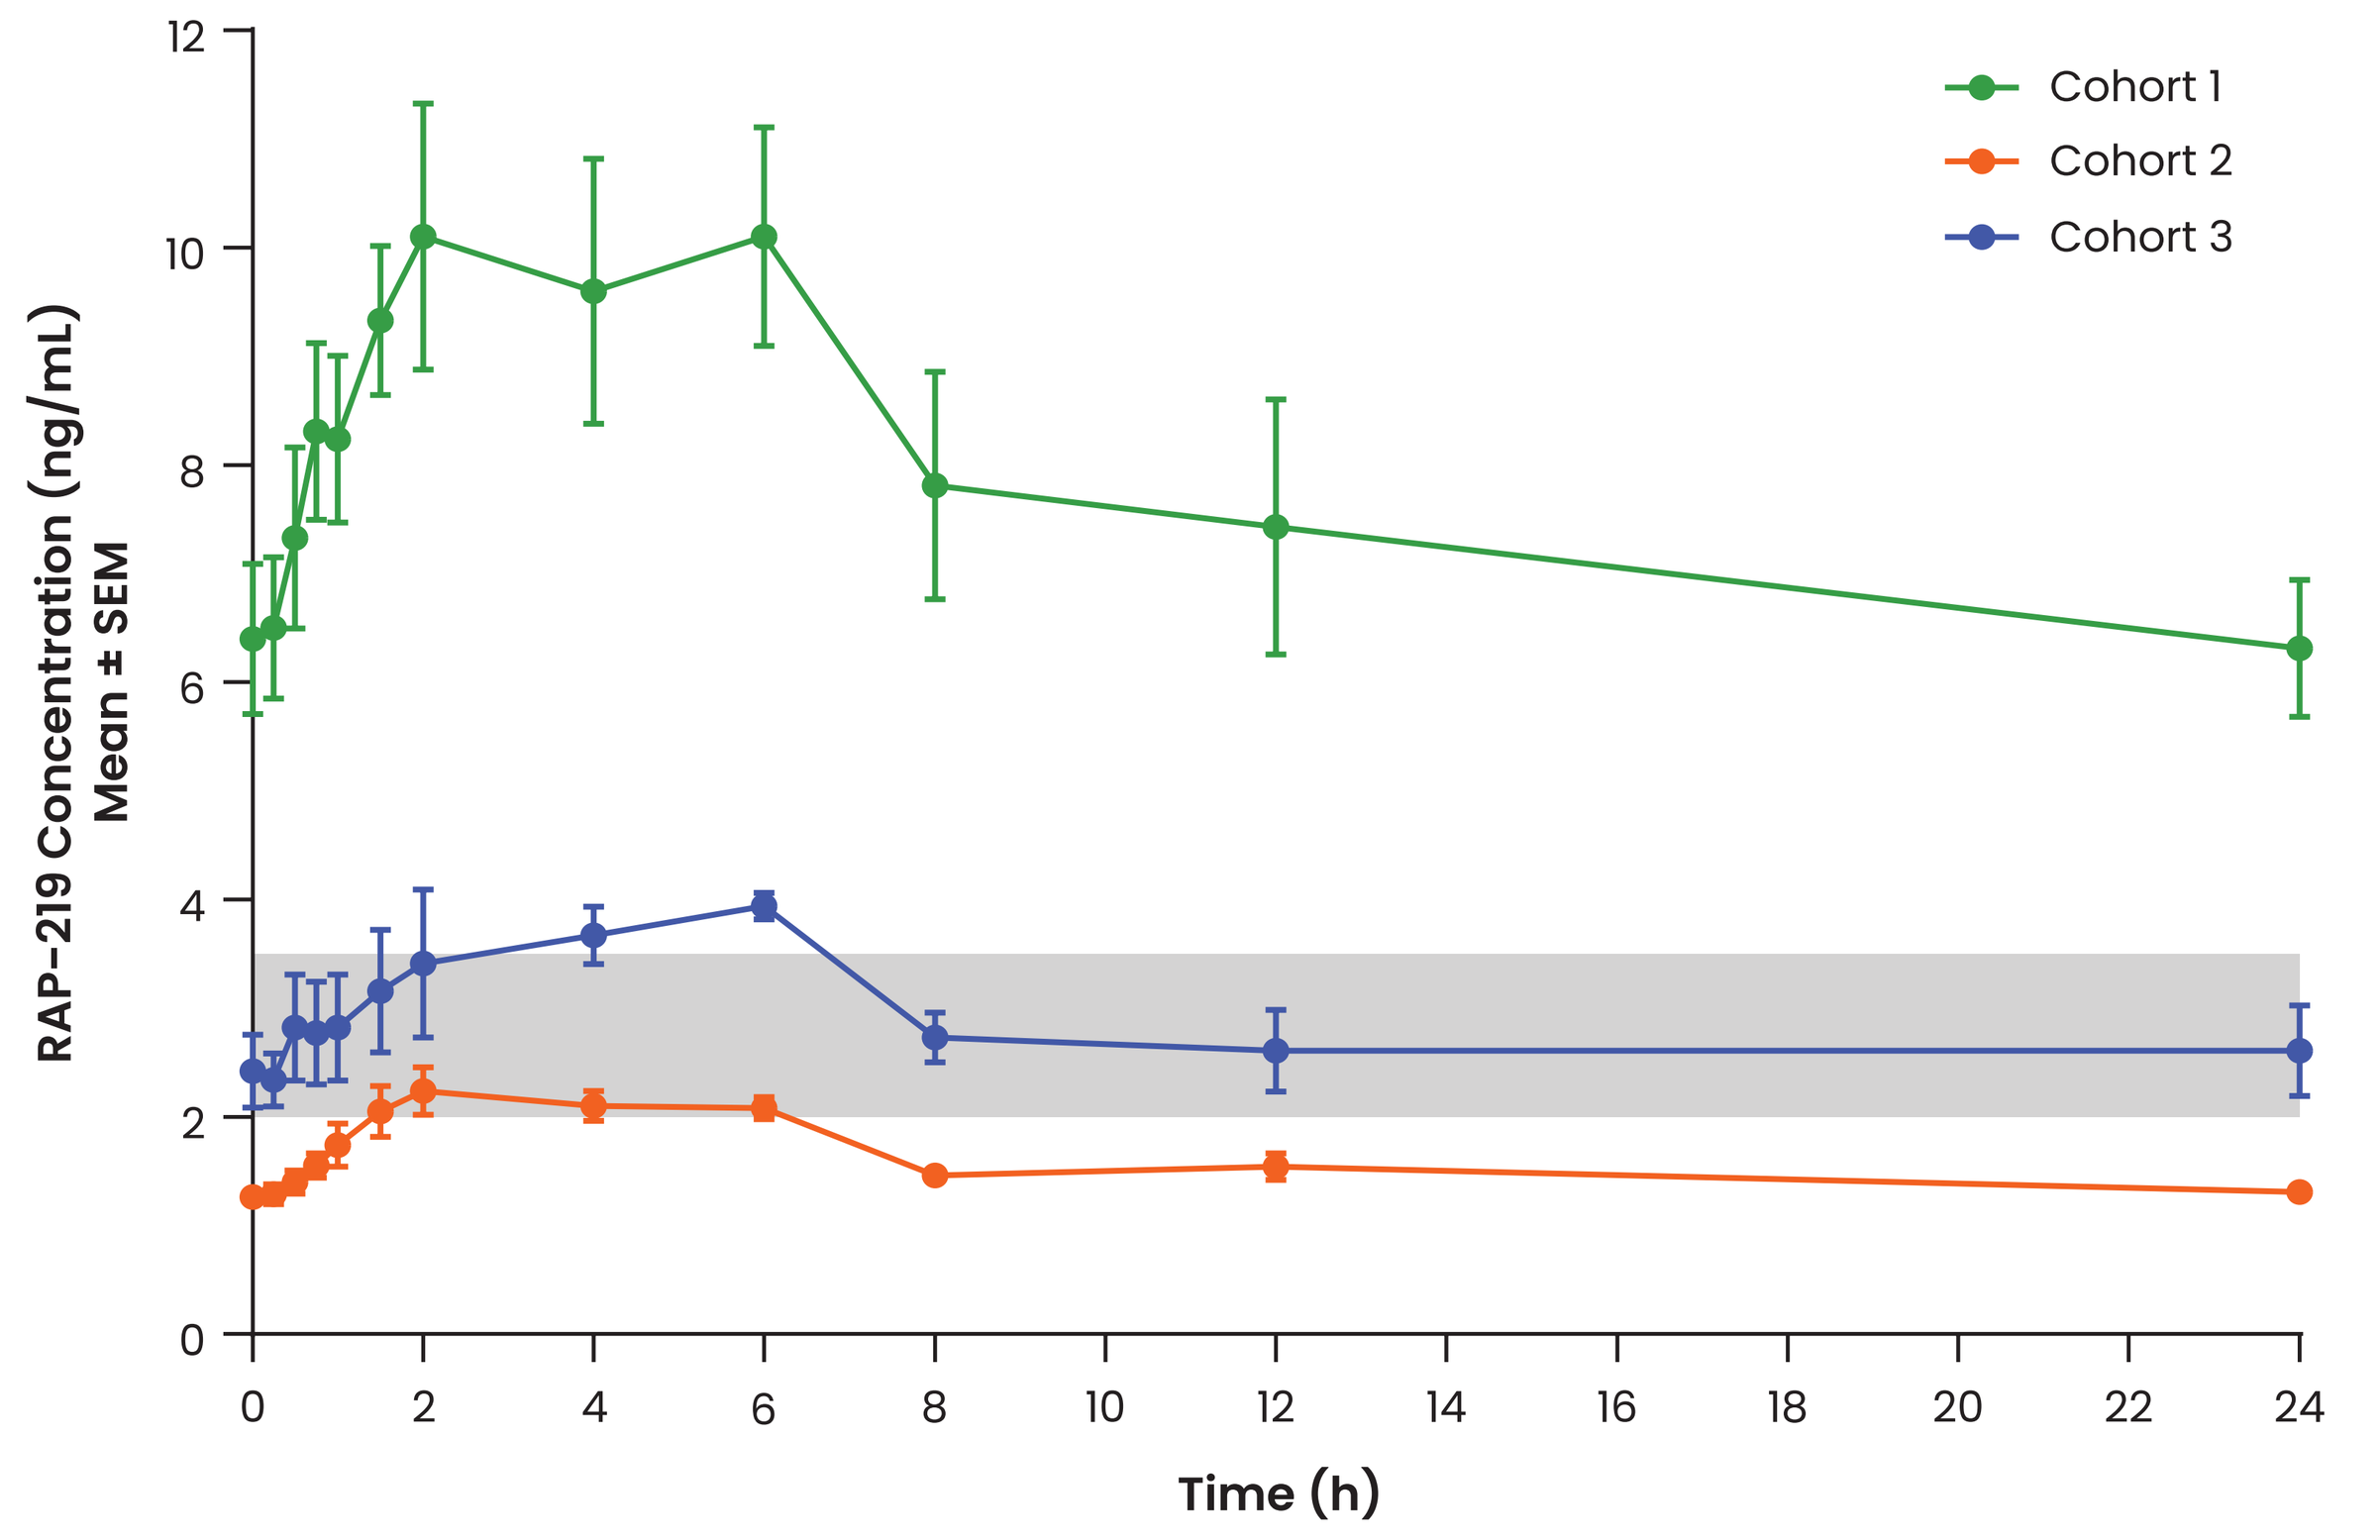
Cohort 1
Cohort 2
Cohort 3
RAP-219 Concentration (ng/mL) Mean ± SEM
Time (h)
0
2
4
6
8
10
12
14
16
18
20
22
24

The shaded region represents the efficacy range (~2-3.5 ng/mL) as estimated by the covalent kinetic model from a preclinical seizure model.
In Cohort 2, two subjects received RAP-219 on D16 instead of D15. The corresponding results were summarized within D15.
SEM – standard error of the mean.

Receptor Occupancy

- Target RO (50%-70%) was achieved with RAP-219 Cp of ~2.0-3.5 ng/mL (**Figure 4**)

Figure 4. Plasma Concentration and Receptor Occupancy of RAP-219

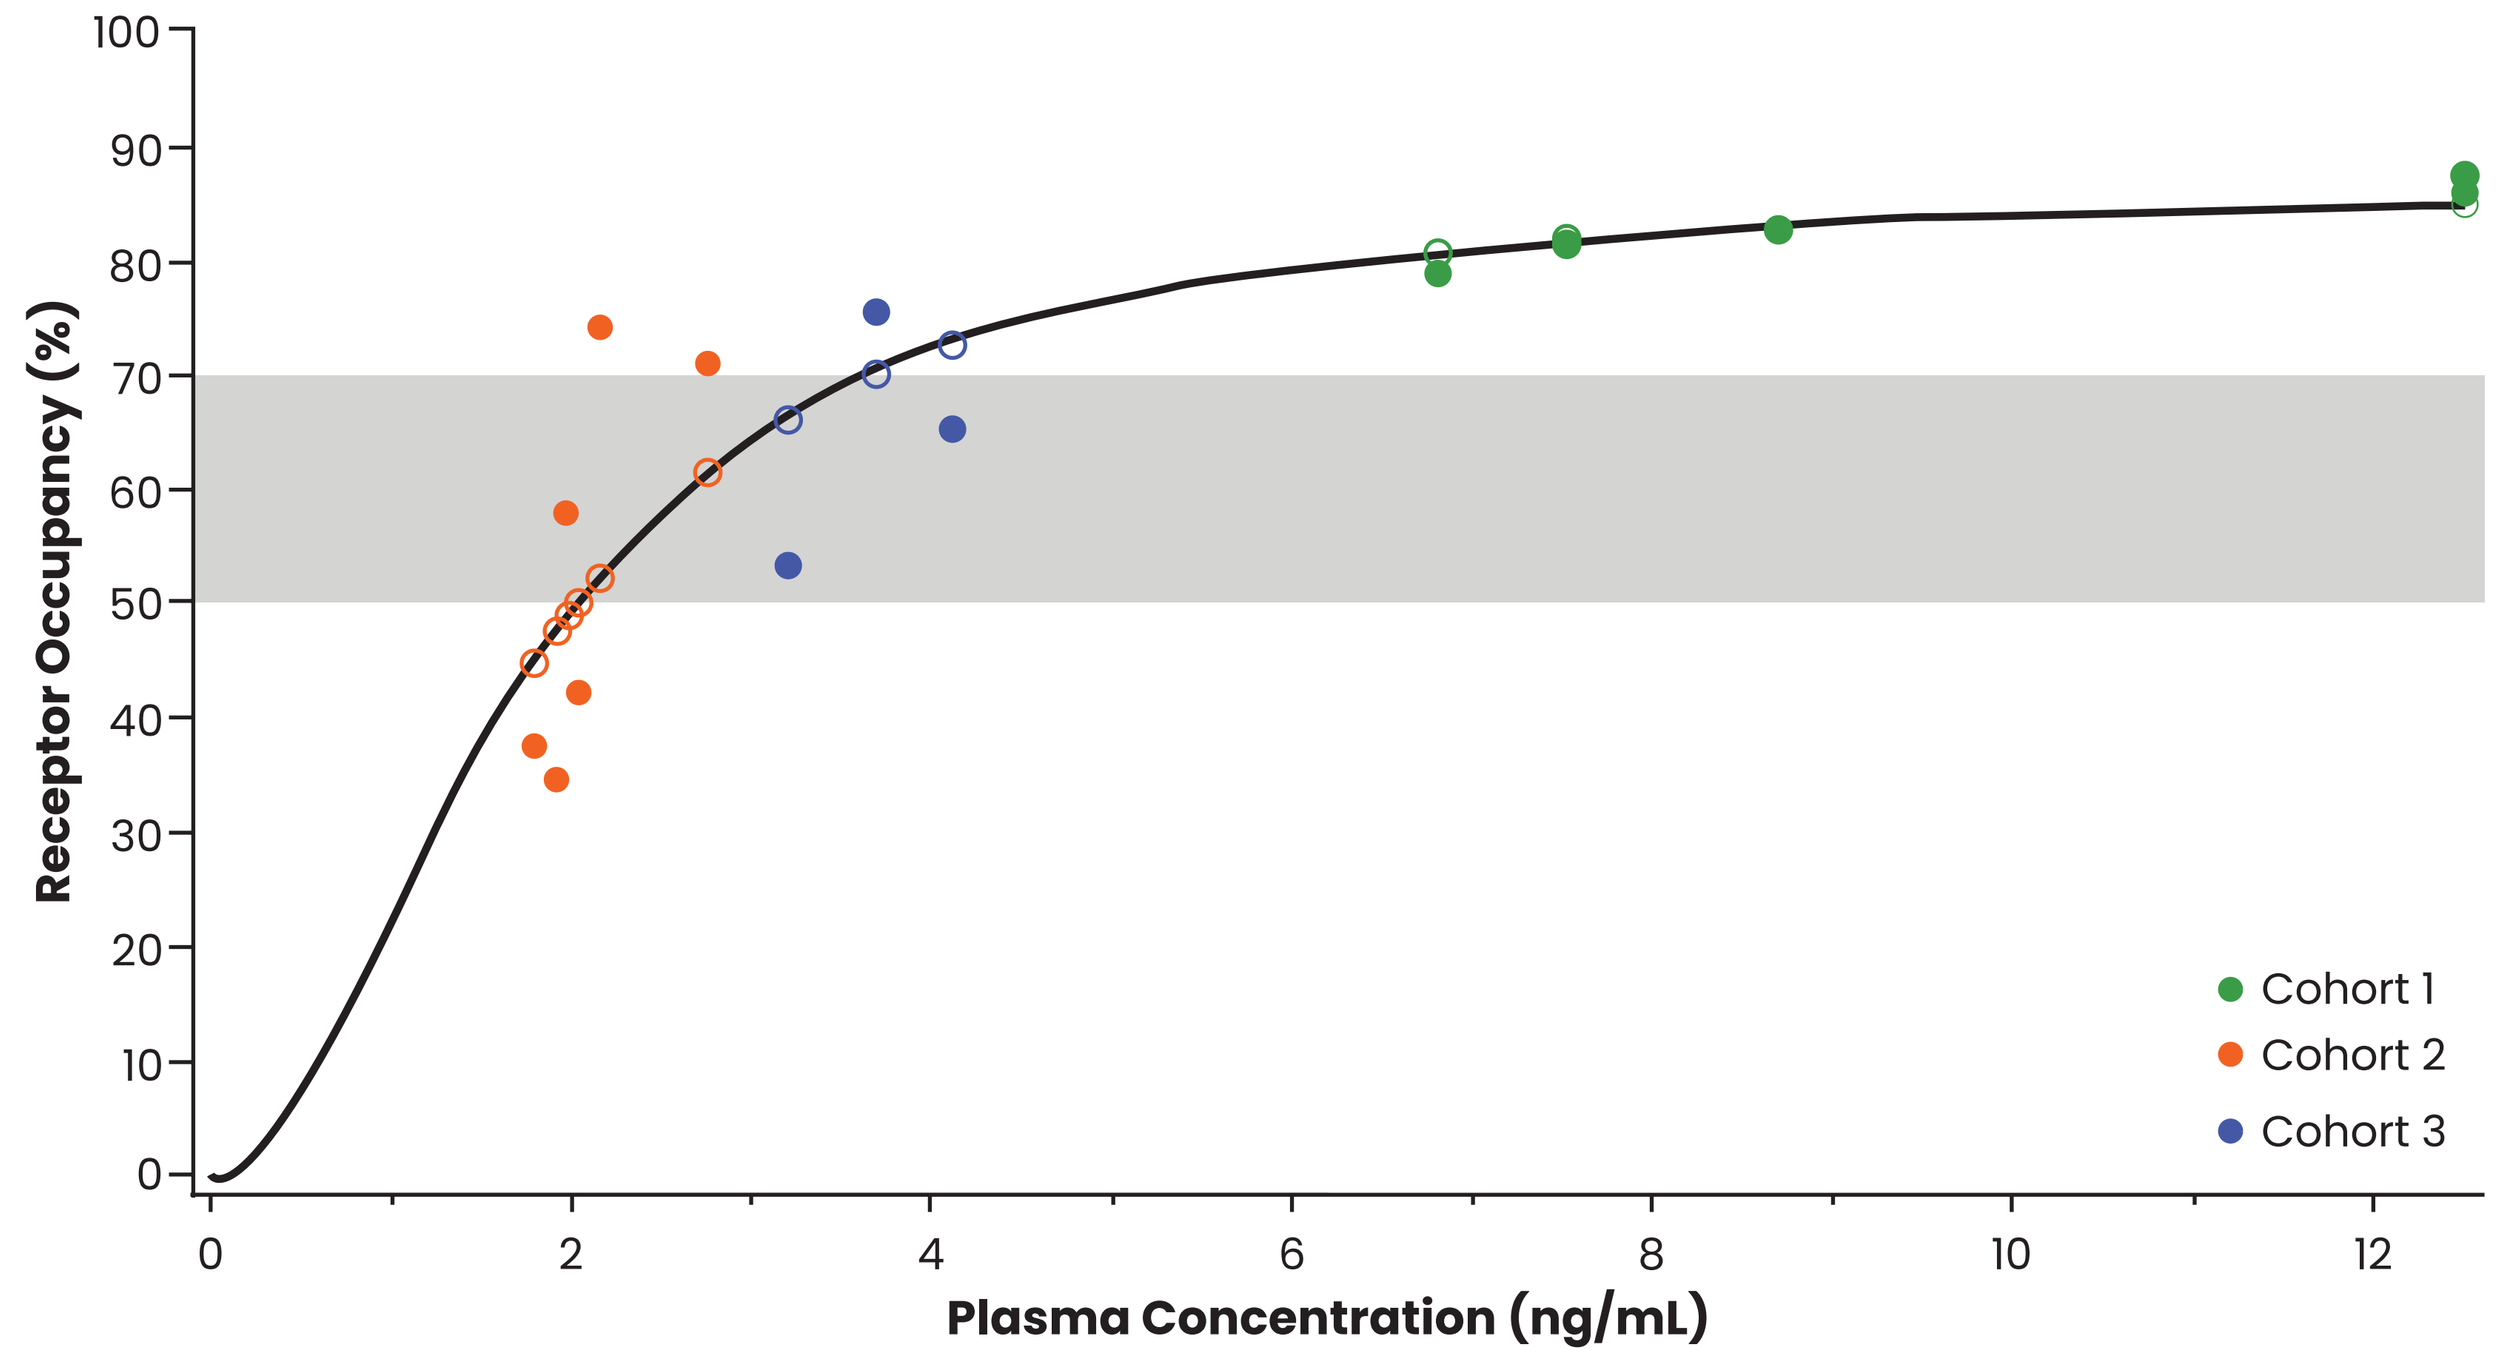
Receptor Occupancy (%)
Plasma Concentration (ng/mL)
Cohort 1
Cohort 2
Cohort 3
0
10
20
30
40
50
60
70
80
90
100
2
4
6
8
10
12

The shaded region represents the target RO associated with maximal efficacy in preclinical seizure models.
Solid data point circles represent actual values; open data point circles represent predicted values.
Cp - plasma concentration; **RO** - receptor occupancy.

Figure 5. Average TARPγ8 3D Volume of Distribution Across Brain Regions at A. Baseline (D1) and B. After RAP-219 Dose (D15)

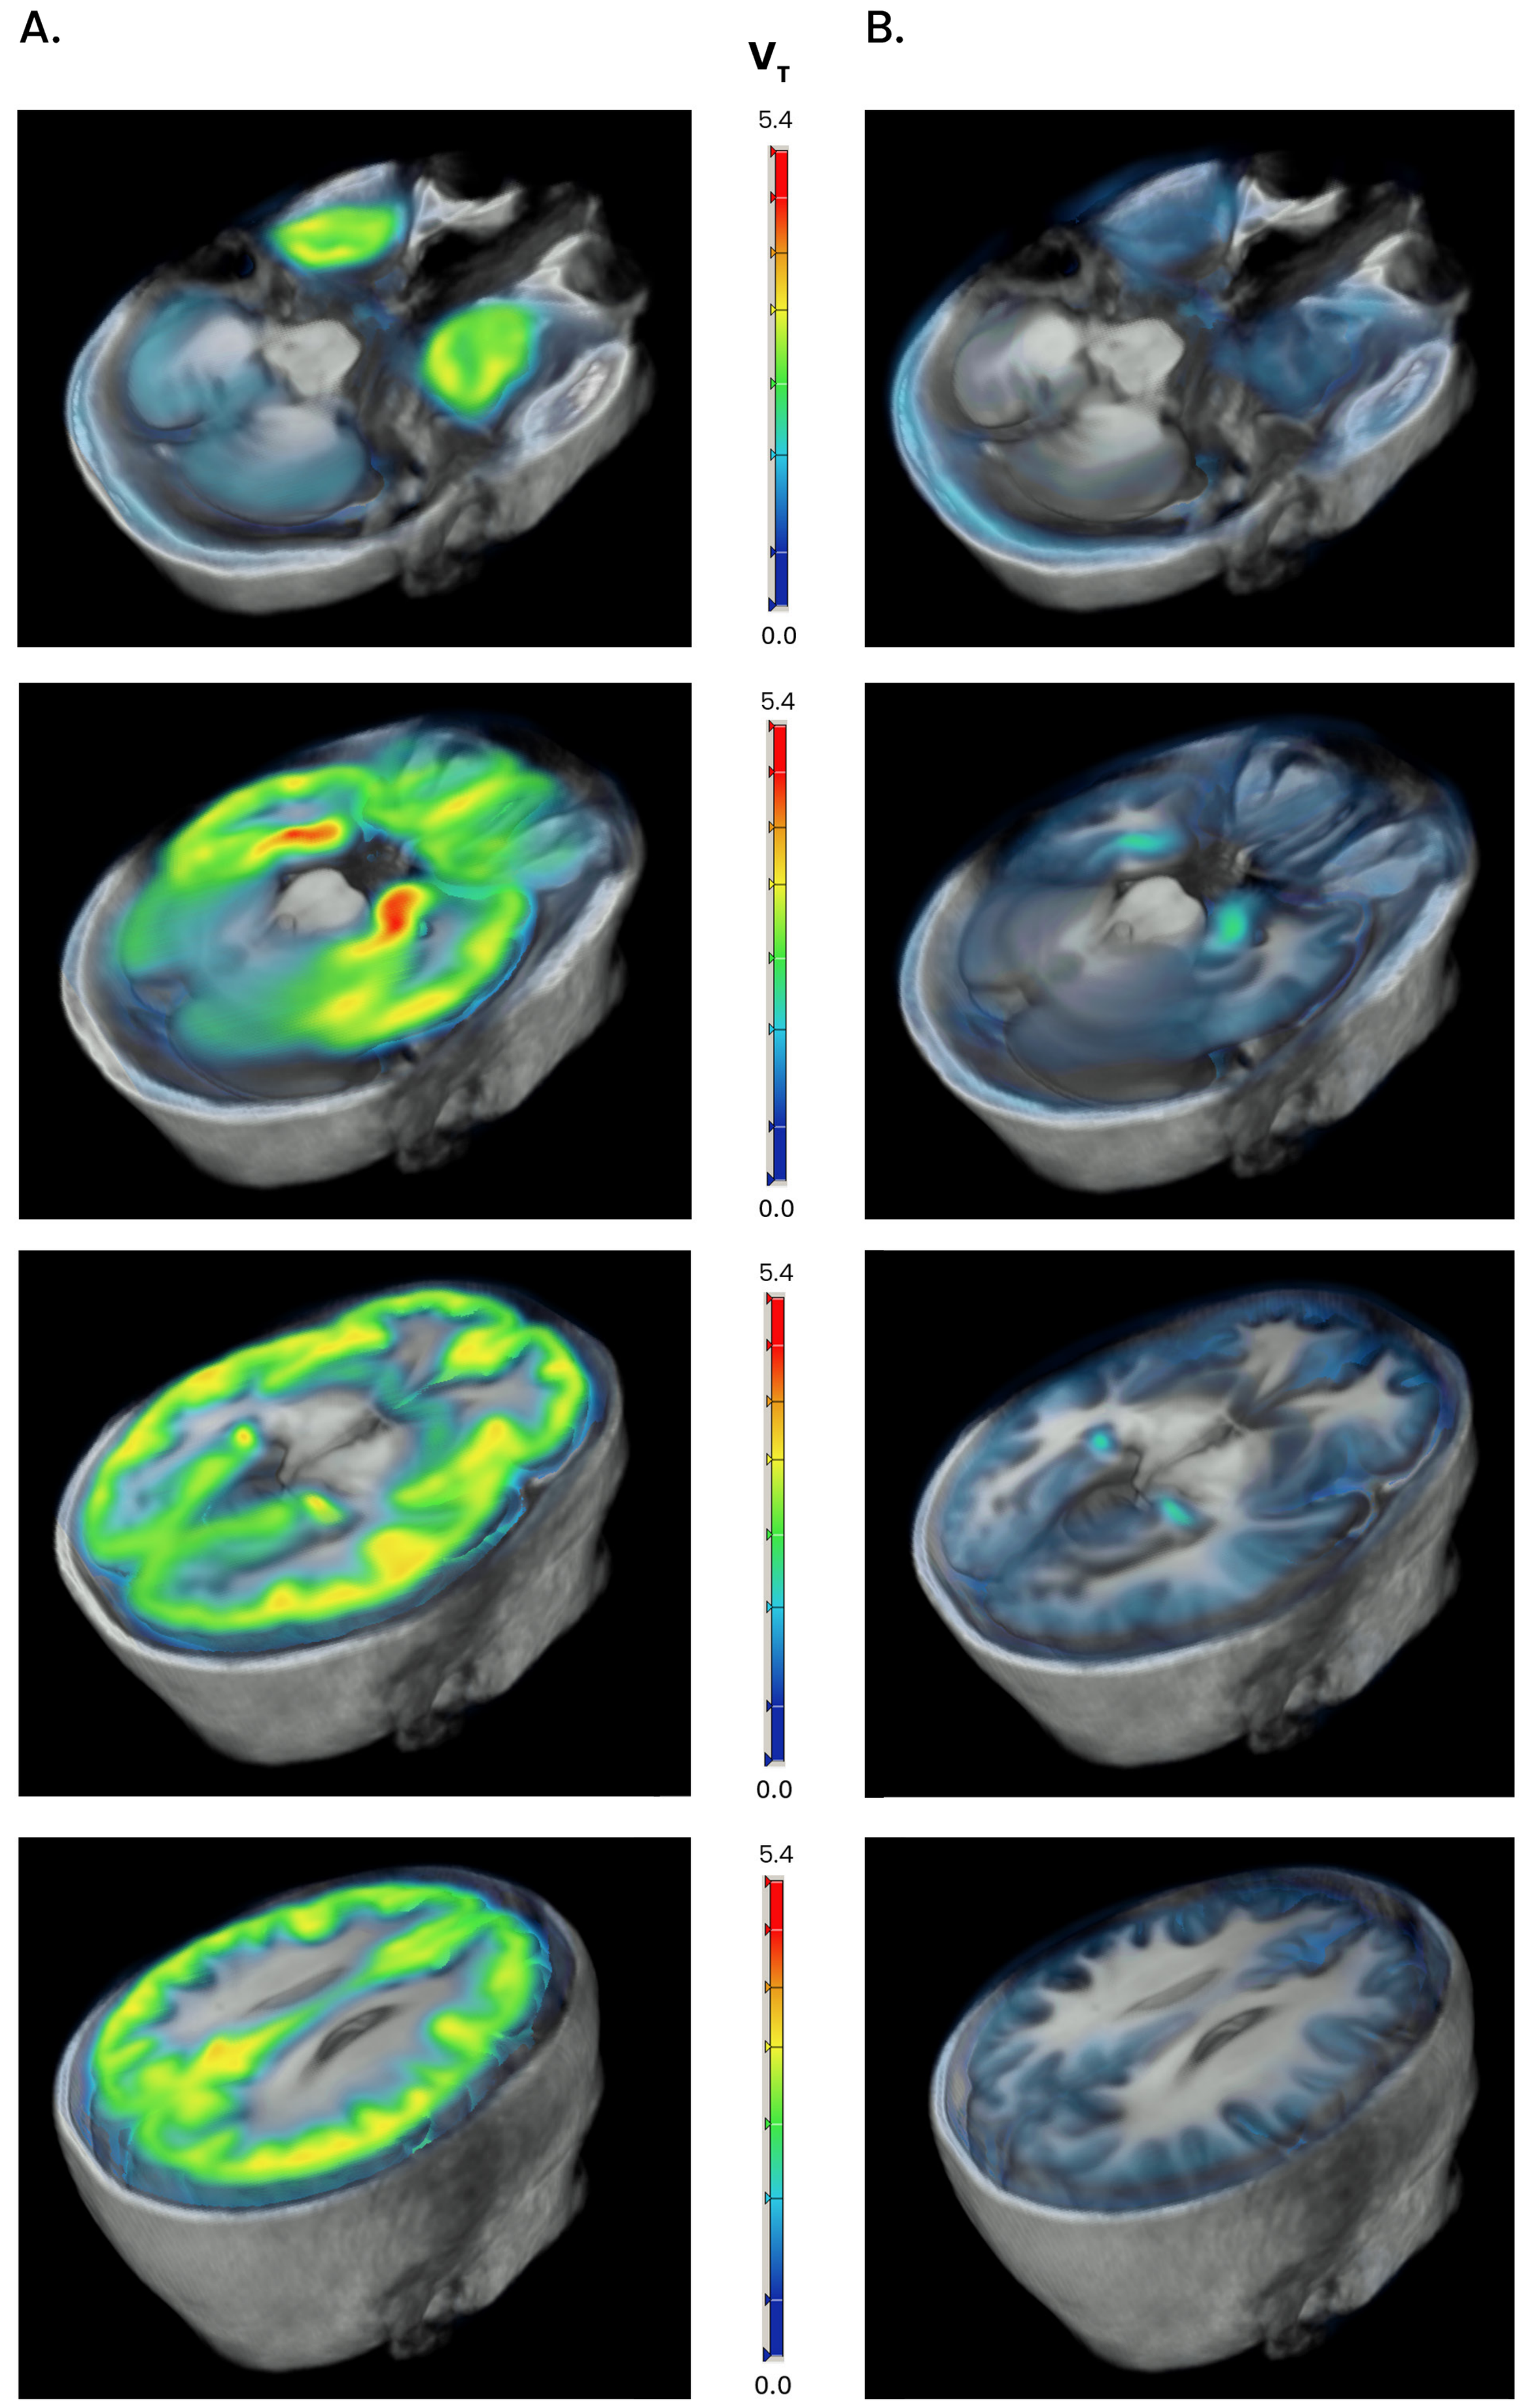
A.
B.
V_T
5.4
0.0

V_T is superposed to a T1-weighted MRI.
3D – 3-dimensional; **MRI** - magnetic responance imaging; V_T – volume of distribution.

Safety and Tolerability

- All treatment-emergent AEs (TEAEs) were Grade 1 (mild) or Grade 2 (moderate)
- One subject from Cohort 1 discontinued due to a treatment-related TEAE (obsessive rumination, Grade 2)

Table 2. Most Common Treatment-Emergent Adverse Events (≥3 Subjects Overall)

	Cohort 1 n=6	Cohort 2 n=6	Cohort 3 n=4	Total N=16
Any TEAE, n (%)	5 (83.3)	5 (83.3)	4 (100)	14 (87.5)
Most Common TEAEs, n (%)				
Headache	2 (33.3)	3 (50)	2 (50)	7 (43.8)
Fatigue	2 (33.3)	2 (33.3)	1 (25)	5 (31.3)
Abnormal dreams	0	0	3 (75)	3 (18.8)
Back pain	0	3 (50)	0	3 (18.8)
Catheter site hematoma	1 (16.7)	2 (33.3)	0	3 (18.8)
Disturbance in attention	1 (16.7)	1 (16.7)	1 (25)	3 (18.8)
Discontinuation due to TEAE, n (%)	1 (16.7)	0	0	1 (6.3)

TEAE – treatment-emergent adverse event.

Conclusions

- This phase 1 PET study confirms the specific neuroanatomical expression of TARPγ8 with enriched expression in the neocortex and MTL (including the amygdala and hippocampus), key regions of the brain involved in the initiation and/or propagation of focal onset seizures (FOS)
 - Minimal expression was observed in the cerebellum, a region of the brain in which AMPA receptor inhibition is associated with CNS side effects such as gait disturbances
- Target RO (50-70%) associated with maximal efficacy in preclinical models was achieved at RAP-219 Cp between ~2.0 and 3.5 ng/mL in human subjects
- The results indicate that doses ≥0.25 mg achieve target RO within 14 days, supporting the use of a range of doses for a phase 3 study in patients with epilepsy
- The RAP-219 PK, safety, and tolerability profiles observed were consistent with those reported in previous phase 1 studies

References

1. Coombs ID, et al. *Mol Pharmacol.* 2022;101(5):343-56.
2. Matta J, et al. American Epilepsy Society (AES) Annual Meeting, Dec 6-10, 2024, Los Angeles, CA, Poster 1.390.

Presented at AES Annual Meeting 2025, December 5-9, 2025, Atlanta, GA.

Acknowledgments

The authors thank Mari Willeman, PhD, and Anthony DiLauro, PhD, of the Sensified division of Woven Health Collective, LLC, for writing and editorial assistance, which were funded by Rapport Therapeutics, Inc.

Disclosures

SG, SY: Rapport, employee and stock ownership.
MC, MK, KS (Kim Serdons), KVL: Nuclear Medicine and Molecular Imaging.
MH, KS (Kathleen Sierens), JDH: Center for Clinical Pharmacology.

Please follow the QR code for a copy of the poster.

Poster ID: 3.355